MP13-2139

A Real-World Analysis of an XLMTM Patient Cohort from the MTM and CNM International Patient Registry

Chiara Marini-Bettolo[1], Ana Buj Bello[2,3], Faryn Solomon[4], Jess Page[1], Tmirah Haselkorn[4], Ioanna Pangou[1], Julie Coats[4], Joanne Bullivant[1], James J. Dowling[5]

[1]The John Walton Muscular Dystrophy Research Centre, Translational and Clinical Research Institute, Newcastle University and Newcastle Hospitals NHS Foundation Trust, Newcastle upon Tyne, UK; [2]Généthon, 91000, Evry, France; [3]Université Paris-Saclay, Univ Evry, Inserm, Généthon, Integrare Research Unit UMR_S951, 91000, Evry, France; [4]Astellas Gene Therapies, San Francisco, CA, USA; [5]The Hospital for Sick Children, Toronto, Canada

Introduction

- Myotubular and other centronuclear myopathies are congenital neuromuscular conditions characterized by central nuclei on muscle biopsy
- Among these, **X-linked myotubular myopathy (XLMTM)** is the most common, affecting ~1 in 40–50,000 newborn males[1]
- XLMTM is characterized by profound muscle weakness leading to impairment of respiratory and neuromuscular function[2,3]
- Half of XLMTM patients do not survive past 18 months of age[2,4]
- There are limited real-world data on the disease burden of XLMTM
- A cross-sectional analysis of the **Myotubular Myopathy (MTM) and Centronuclear Myopathy (CNM) Patient Registry** was performed to characterize individuals with XLMTM in a real-world setting

The Myotubular and Centronuclear Myopathy Patient Registry

About the MTM and CNM Patient Registry https://mtmcnmregistry.org/

- International, disease-specific, longitudinal, open-ended database, founded in 2013
- Operated by the John Walton Muscular Dystrophy Research Centre at Newcastle University (UK), and affiliated to the TREAT-NMD network
- Patient- or caregiver-initiated registration through an online portal
- Information reported by the patient or caregiver and their nominated clinician
- Demographic, genetic, and clinical data from individuals with a diagnosis of MTM or CNM, carrier females of XLMTM, and deceased individuals with a confirmed diagnosis
- The registry is governed by an independent expert steering committee

Objective

- To describe demographic and clinical characteristics of living male patients with XLMTM in the MTM and CNM Patient Registry, with a focus on healthcare utilization, motor and respiratory function, and family history

Methods

Study Design

- Cross-sectional analysis of a living male patient cohort with genetically confirmed XLMTM from the MTM and CNM Patient Registry; data lock was on July 22, 2022

Data analysis

- Results were de-identified and reported at the aggregate level from patients' most recent data entry
- As data in the registry were patient-entered, there were varying response rates for certain questions; denominators reported in the results represent the number of reliable responses for each outcome; analyses were not performed for response rates of ≤2 patients
- For analyses of average values, the mean was provided where n>10 and the median where n<10, unless stated otherwise

Results

Demographic characteristics

- Participants: 88 male patients with genetically confirmed XLMTM from 25 countries (**Fig. 1**)
- Mean age (± SD) at genetic diagnosis: 4.1 ± 8.9 years (range 0–46)
- Mean age (± SD) at data cutoff date: 11.7 ± 11.7 years (range 0–59)

Figure 1. Number of patients by country

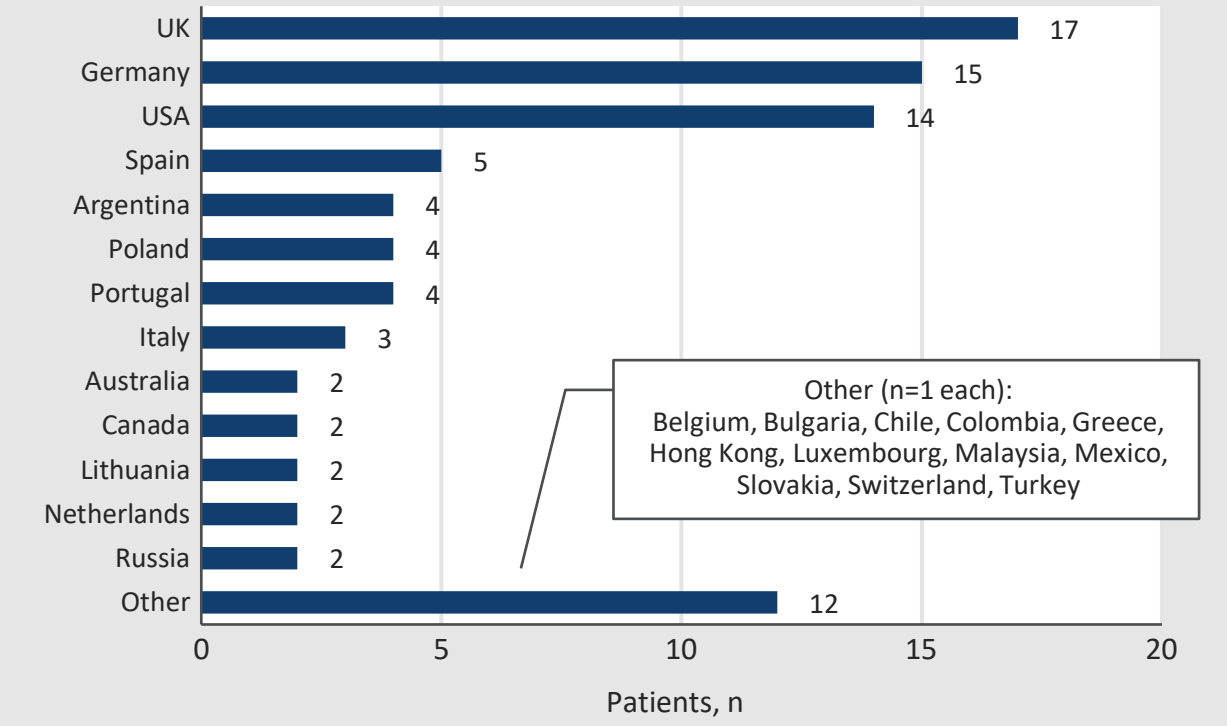
UK 17
Germany 15
USA 14
Spain 5
Argentina 4
Poland 4
Portugal 4
Italy 3
Australia 2
Canada 2
Lithuania 2
Netherlands 2
Russia 2
Other 12
0 5 10 15 20
Patients, n
Other (n=1 each):
Belgium, Bulgaria, Chile, Colombia, Greece, Hong Kong, Luxembourg, Malaysia, Mexico, Slovakia, Switzerland, Turkey

Figure 2. Measures of healthcare utilization and disease burden

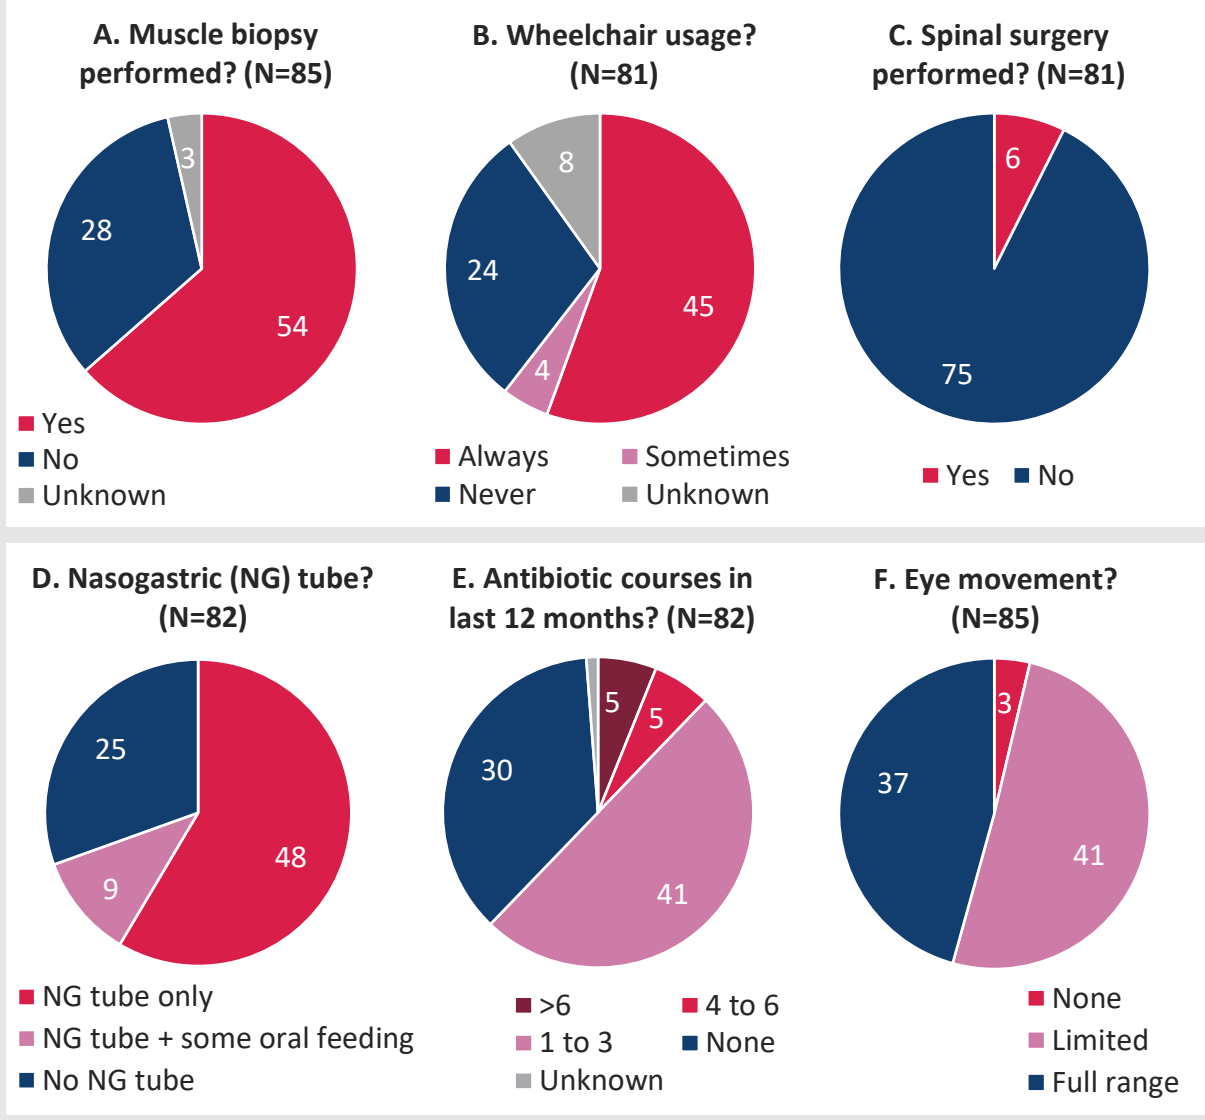
A. Muscle biopsy performed? (N=85)
54
28
3
Yes
No
Unknown
B. Wheelchair usage? (N=81)
45
24
8
4
Always
Never
Sometimes
Unknown
C. Spinal surgery performed? (N=81)
6
75
Yes
No
D. Nasogastric (NG) tube? (N=82)
48
25
9
NG tube only
NG tube + some oral feeding
No NG tube
E. Antibiotic courses in last 12 months? (N=82)
5
5
41
30
>6
4 to 6
1 to 3
None
Unknown
F. Eye movement? (N=85)
3
41
37
None
Limited
Full range

Healthcare Utilization / Disease Burden

- 54/85 patients (63.5%) had received a muscle biopsy (**Fig. 2A**)
- 45/81 patients (55.6%) had permanent reliance on wheelchair use (**Fig. 2B**); based on responses from 13 of these patients, the mean age at which wheelchair dependence started was 67 months
- Spinal surgery for scoliosis was performed in 6/81 (7.4%) patients (**Fig. 2C**), at a median age of 118 months (i.e., 9.8 years)
- 48/82 (58.5%) could feed only through a gastric or nasal tube (**Fig. 2D**)

Attainment of Motor Milestones

- Among patients with reliable data for best motor milestone achievement, 33/79 (42%) were unable to sit or walk without support at any time (**Fig. 3**)
- Reported current milestones achieved indicate overall worsening in motor function compared to best milestones achieved (**Fig. 3**)

Respiratory Function

- Of 88 patients, most (73.9%) reported requiring ventilation at birth
- 44/88 (50.0%) currently use invasive ventilation, 31.8% use non-invasive ventilation, 11.4% use no ventilation, and 6.8% did not specify
- 40/88 (45.5%) currently use ventilation for ≥16 hours/day (**Fig. 4**)
- 51/82 (62.2%) required antibiotics for chest infections on between 1 and >6 occasions in the past 12 months (**Fig. 2E**)

Figure 3. Best and current motor milestones achieved

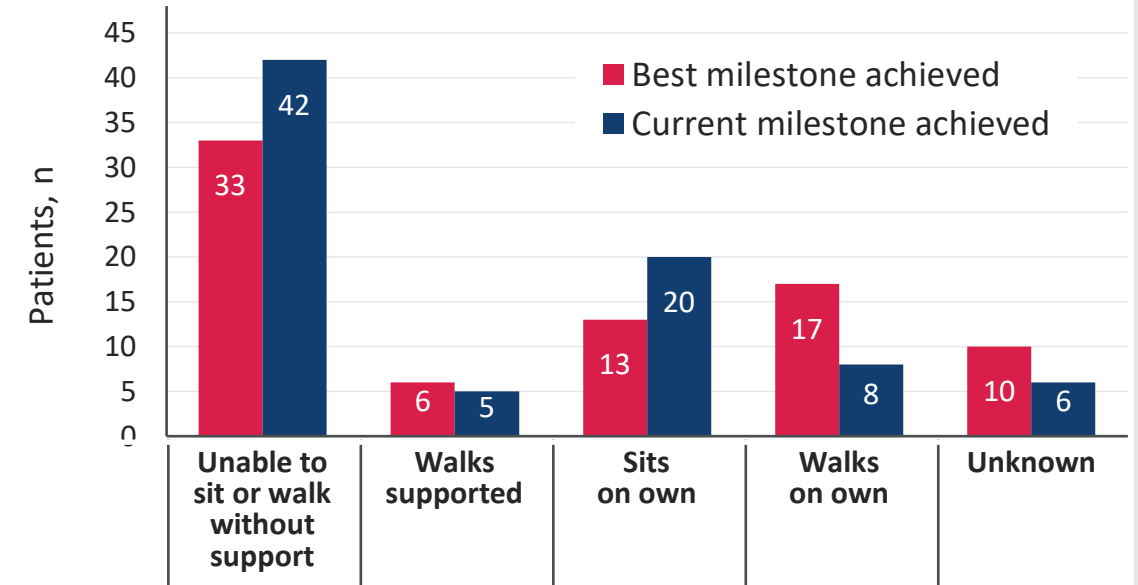
Patients, n
0 5 10 15 20 25 30 35 40 45
Best milestone achieved
Current milestone achieved
33 42
6 5
13 20
17 8
10 6

	Unable to sit or walk without support	Walks supported	Sits on own	Walks on own	Unknown
Best milestone, n (%)[a]	33 (41.8%)	6 (7.6%)	13 (16.5%)	17 (21.5%)	10 (12.7%)
Age[b] achieved from (months)	n/a	26 (n=4)	22.5 (n=11)	23.4 (n=15)	n/a
Age[b] achieved until (months)	n/a	120 (n=3)	– (n=2)	138 (n=8)	n/a
Current milestone, n (%)[c]	42 (51.85%)	5 (6.2%)	20 (24.7%)	8 (9.9%)	6 (7.4%)
Age[b] achieved from (months)	156 (n=3)[d]	134 (n=5)	42 (n=18)	18 (n=6)	n/a

[a]N=79; [b]Average calculated as mean where n>10 and median where n<10; [c]N=81; [d]i.e. 3 patients had previously had a better motor milestone but lost this function and became 'unable to sit or walk without support' at median age of 156 months. 'n' denotes the number of reliable responses available for each analysis. n/a, not applicable.

Other Clinical Parameters

- 41/81 (50.6%) and 3/81 (3.7%) patients reported limited or no ability to move their eyes (**Fig. 2F**)
- Cardiac manifestations affected fewer patients: 10/82 (12.2%) and 7/82 (8.5%) reported abnormal echocardiogram (ECHO) or electrocardiogram (ECG) results, respectively

Figure 4. Ventilator dependence

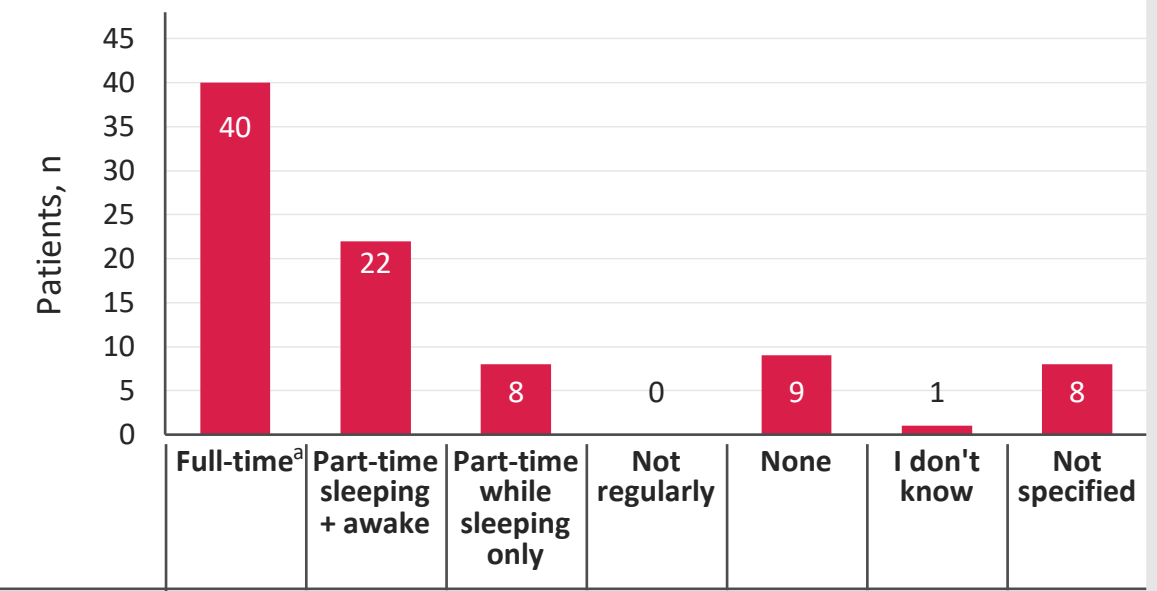
Patients, n
0 5 10 15 20 25 30 35 40 45
40
22
8
0
9
1
8

	Full-time[a]	Part-time sleeping + awake	Part-time while sleeping only	Not regularly	None	I don't know	Not specified
	If ventilation is used, from what age (months) has it been required?						
Mean/median[b] [range] (n)	23 [0–297] (n=36)	31 [0–180] (n=21)	16 [0–173] (n=8)	n/a	n/a	n/a	n/a

[a]At least 16 hours per 24 hours; [b]Average calculated as mean where n>10 and median where n<10. 'n' denotes the number of reliable responses available for each analysis. n/a, not applicable.

Conclusions

- The MTM-CNM Patient Registry provides a unique opportunity to examine real-world data in patients with XLMTM
- The disease burden of XLMTM is substantial, with most patients experiencing limited motor function and requiring respiratory support at birth and in daily life
- Use of assistive devices and gastric or nasal tubes was common
- Limitations include varying response rates, missing data, and the cross-sectional nature of this analysis
- These data contribute to the understanding of XLMTM and highlight the need for improved treatments for these patients

ABBREVIATIONS CNM, centronuclear myopathy; MTM, myotubular myopathy; SD, standard deviation; XLMTM, X-linked myotubular myopathy.

REFERENCES **1.** Vandersmissen I, et al. Neuromuscul Disord 2018;28:766–67; **2.** Beggs AH, et al. *Muscle Nerve* 2018;57: 550–60; **3.** Jungbluth H, et al. *Orphanet J Rare Dis* 2008;3:26; **4.** McEntagart M, et al. Neuromuscul Disord 2002;12: 939–46.

DISCLOSURES **CMB, JP, IP, and JB** have received study funding related to this presentation from Astellas, the Myotubular Trust, and Muscular Dystrophy UK. **ABB** has received consulting fees from Audentes Therapeutics (now Astellas Gene Therapies) and research funding from the Myotubular Trust; she also holds a patent on systemic gene replacement therapy for treatment of XLMTM. **JJD** has received research grants or contracts from Astellas Gene Therapies for clinical trials and preclinical studies relating to gene therapy in XLMTM; and has received an honorarium for a sponsored symposium and support for travel to an international meeting to present data from Astellas Gene Therapies. **FS and TH** are employees and shareholders of Astellas Gene Therapies. **JC** is an employee of Astellas Gene Therapies. The Myotubular and Centronuclear Myopathy Patient Registry was developed in partnership with the MTM and CNM communities and is jointly funded by the Myotubular Trust, Muscular Dystrophy UK, and Astellas Pharma, Inc.
Fourwave Medical Communications provided editorial support and poster layout, funded by Astellas Gene Therapies.

Presented at the 15th Congress of the European Paediatric Neurology Society (EPNS), 20–24 June 2023, Prague, Czech Republic

©(2023) by Astellas and the Authors. Contents not to be distributed, reproduced, presented, or otherwise shared without permission. All rights reserved